Assay Interference and Blood Transfusion Safety in Patients With Relapsed or Refractory Multiple Myeloma (MM) Treated With Daratumumab

41

Donna Catamero, ANP-BC, OCN, CCRC[1,*]; Gillian Morgan, RN, BSN, CCRP[2]; Imran Khan, MD, PhD[3]

[1]The Mount Sinai Medical Center, New York, NY; [2]Mount Sinai Hospital School of Medicine, New York, NY; [3]Janssen Research & Development, LLC, Raritan, NJ.

*Presenting author.

INTRODUCTION

- Daratumumab (DARA) is a first-in-class, human IgG1 monoclonal antibody that targets CD38,[1] a protein that is highly expressed on multiple myeloma (MM) cells[2,3]
 - DARA was recently approved by the US Food and Drug Administration for use in patients with heavily pretreated MM (>3 prior lines of therapy) or patients with MM that is refractory to a proteasome inhibitor (PI) and an immunomodulatory drug (IMiD)[4]
- In 2 clinical studies of DARA monotherapy (16 mg/kg) in patients with relapsed or refractory MM (GEN501 [ClinicalTrials.gov Identifier: NCT00574288] and SIRIUS [NCT01985126]), overall response rates were 36% and 29%, respectively, and included complete responses (CRs) and stringent CRs (sCRs)[5,6]
- In the relapsed and refractory treatment setting, patients with MM frequently have anemia and often require blood transfusions
- During routine blood bank screening during the GEN501 study, unexpected positive indirect Coombs tests in DARA-treated patients were reported[7,8]
 - Indirect Coombs tests are often performed to detect alloantibodies in recipient and donor blood before a red blood cell (RBC) transfusion
- CD38 is also expressed on human RBCs, and DARA may interfere with indirect Coombs tests by binding to CD38 on RBCs (**Figure 1**)

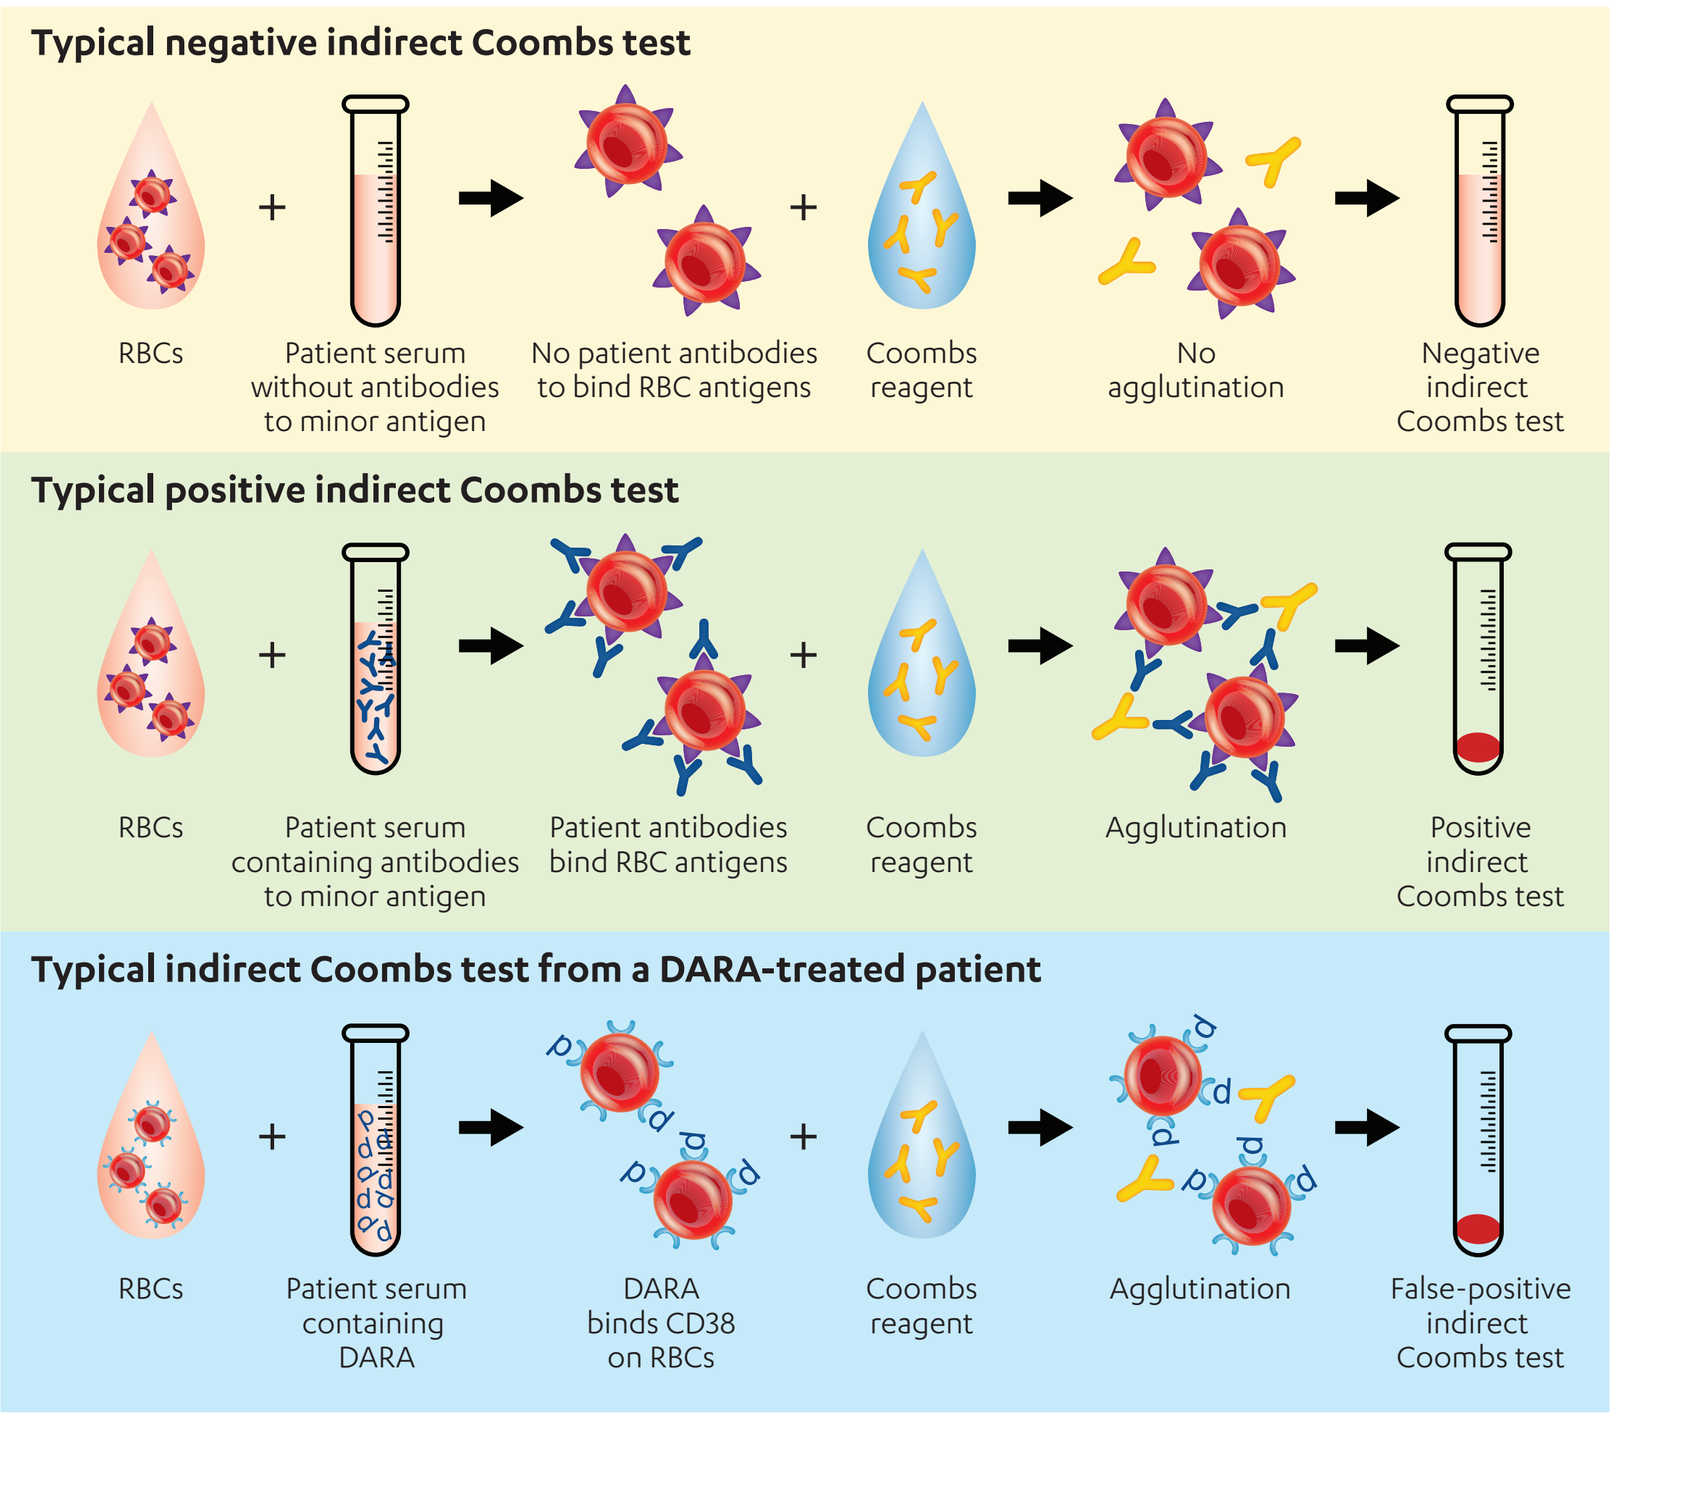
Typical negative indirect Coombs test
RBCs
Patient serum without antibodies to minor antigen
No patient antibodies to bind RBC antigens
Coombs reagent
No agglutination
Negative indirect Coombs test
Typical positive indirect Coombs test
RBCs
Patient serum containing antibodies to minor antigen
Patient antibodies bind RBC antigens
Coombs reagent
Agglutination
Positive indirect Coombs test
Typical indirect Coombs test from a DARA-treated patient
RBCs
Patient serum containing DARA
DARA binds CD38 on RBCs
Coombs reagent
Agglutination
False-positive indirect Coombs test

DARA, daratumumab; RBC, red blood cell.

Figure 1. Mechanism of DARA interference with indirect Coombs tests.

- DARA assay interference may cause a delay in the release of blood products from blood blanks
 - One strategy to negate DARA interference is to treat RBCs with dithiothreitol (DTT) to denature CD38 so that DARA no longer binds to RBCs[7]
 - Alternatively, RBC phenotyping may be initiated prior to the first DARA dose
- Here, we present an analysis of outcomes in patients at Mount Sinai who received RBC transfusions while participating in the SIRIUS study, and highlight the role of oncology nurses in mitigating the effects of DARA interference in blood typing assays

OBJECTIVES

- To present one clinical center's experience with managing blood transfusions in DARA-treated patients
- To highlight the role of oncology nurses in mitigating interference with indirect Coombs tests in order to facilitate safe, efficient delivery of blood products to patients

METHODS

Study Design

- SIRIUS was an open-label, 2-part, international, multicenter, phase 2 study
- In Part 1, patients were randomized 1:1 to receive either DARA 16 mg/kg weekly for 8 weeks, then every 2 weeks for 16 weeks, and every 4 weeks thereafter, or DARA 8 mg/kg every 4 weeks (**Figure 2**)

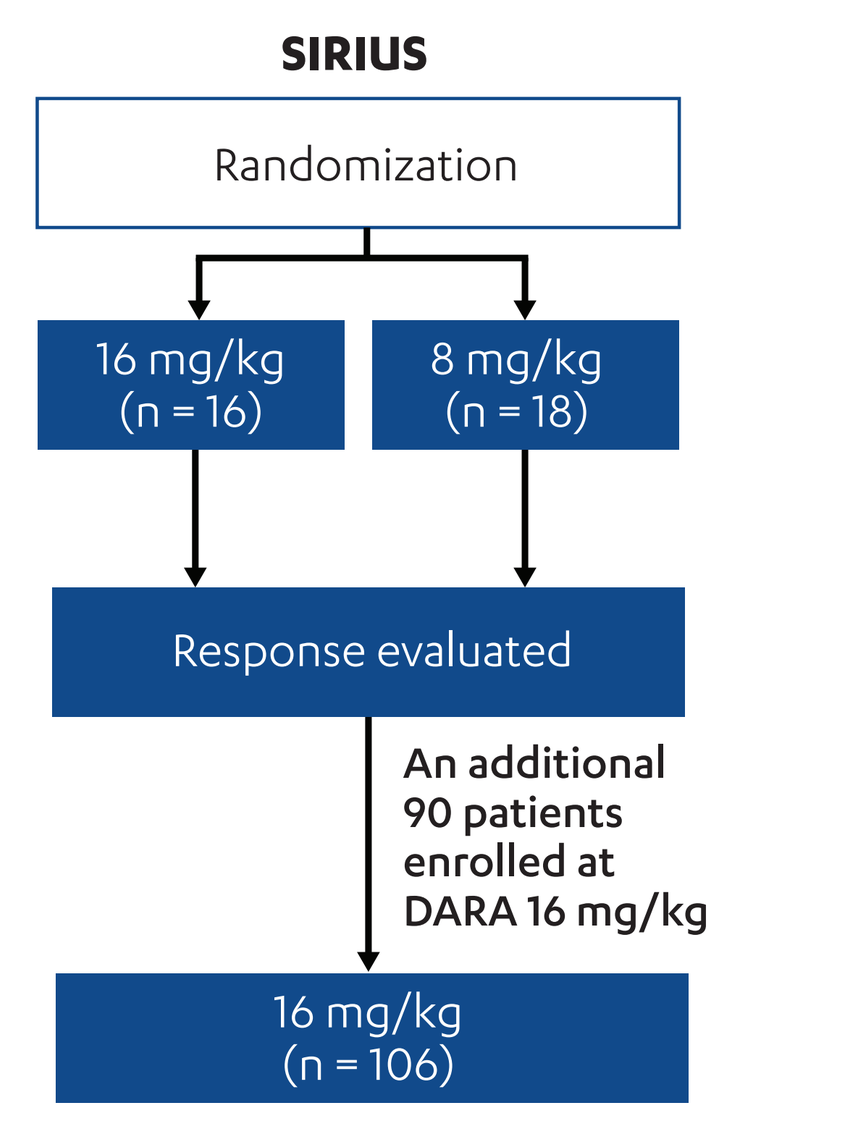
SIRIUS
Randomization
16 mg/kg (n = 16)
8 mg/kg (n = 18)
Response evaluated
An additional 90 patients enrolled at DARA 16 mg/kg
16 mg/kg (n = 106)

DARA, daratumumab.

Figure 2. SIRIUS study design.

- Based on an interim analysis, the recommended dose (DARA 16 mg/kg) and dose schedule were established and selected for further study in Part 2
- Throughout the study, any treatments that were deemed necessary to provide adequate supportive care, including blood transfusions, were prescribed by investigators

Eligibility Criteria

- Documented MM and evidence of disease progression on or within 60 days of the most recent prior treatment regimen
- Evidence of response to ≥1 prior treatment regimen
- Received ≥3 prior lines of therapy, including a PI and an IMiD, in any order during the course of treatment, or disease was double refractory to a PI and an IMiD
- Eastern Cooperative Oncology Group performance status of 0 to 2
- Patients who received prior allogeneic stem cell transplantation were excluded; patients could not have received autologous stem cell transplantation within 12 weeks before the first treatment cycle
- Patients with nonsecretory MM based upon standard M-component criteria were excluded, unless the baseline serum free light chain level was elevated

Blood Typing

- RBC phenotyping by tube methodology was routinely performed prior to patients receiving the first dose of DARA
- If the patient's antibody screen was positive, antibody identification was performed using a panel of reagent RBCs
- Antibody screens were performed in gel cards using an automated ProVue® analyzer (Ortho Clinical Diagnostics, Raritan, NJ)
 - RBC panels, including an autocontrol that tested a patient's RBCs against their own plasma, were performed using gel cards
- If panreactivity was detected on gel cards, samples were manually tested further using low ionic strength saline (LISS)

Change in Hemoglobin Levels Post-transfusion

- The change in hemoglobin (Hb) per unit was calculated as the difference between the pre-transfusion value and a reading within 1 week of the transfusion date. If >1 post-transfusion value was recorded within 1 week, the mean of the differences per unit was reported

RESULTS

Transfusions in the Overall Study Population

- At a clinical cut-off date of January 9, 2015, 42 (40%) patients receiving DARA 16 mg/kg required transfusions during treatment, some of whom received both RBC and platelet transfusions
 - 40 (38%) patients received a total of 126 transfusions of packed RBCs
 - 14 (13%) patients received a total of 67 transfusions of platelets
- Thus far, only 1 transfusion-related reaction (grade 1 hives, itching, and facial flush) has been observed
 - The reaction occurred following platelet, not RBC, transfusion and was deemed unrelated to DARA

Transfusion Experience at Mount Sinai

Blood Typing

- Eight patients were enrolled at our institution and commenced treatment between March 4, 2014 and May 20, 2014[9]
 - Antibody screening by indirect Coombs test was performed on 7 patients prior to receiving their first dose of DARA
 - Two patients showed multiple alloantibodies, including anti-D, anti-E, anti-K, anti-Jkb, anti-Fya, anti-Fyb, anti-S, and anti-Knops (**Table 1**)
 - These patients and 5 others agglutinated all RBCs on panel testing, with weak reactivity at the antihuman globulin phase of testing
 - Six of these 7 patients showed a negative autocontrol. One patient with a positive autocontrol had a weakly positive direct antiglobulin test with IgG
 - Six of these 7 patients with panagglutinin showed a positive result on the antibody screen and on the antibody identification panel after initiation of DARA
 - ◊ This positivity continued for 7 to 175 days (median, 49 days)
 - One patient did not have an indirect Coombs test after initiation of DARA

Blood Transfusions

- A total of 9 leukoreduced, irradiated, phenotypically matched RBCs were given to 3 patients during DARA treatment (**Table 2**)
- An additional 9 units were given to 3 patients after DARA completion, while their indirect Coombs tests remained positive
- All transfusions resulted in an appropriate rise in Hb (median, 1.0 g/dL [range, 0.5-2.7])
- No associated transfusion reactions were observed
- None of the patients made new, unexpected RBC alloantibodies while receiving phenotypically matched RBCs

Table 1. Blood Typing of Patients

Patient ID	RBC antigen antibody	Strength of panhemagglutinin	Additional comments
1	–	2+ (gel), 1+ (LISS)	AC negative, not enhanced by ficin
2	–	2+ (gel), 1+ (LISS)	AC1+ (gel), AC– (LISS), DAT IgG1+, eluate– (gel)
3	Anti-D and anti-E	1-2+ (gel), no reaction (LISS)	AC negative
4	–	1+ (gel), 0-1 (LISS)	AC negative
5	Anti-E, anti-K, anti-Jkb, anti-Fya, anti-Fyb, anti-S, and anti-Knops	2+ (gel), 1+ (LISS)	AC negative
6	N/A	N/A	N/A
7	–	2-3+ (gel), 1+ (LISS)	AC negative, negative at IS and 37
8	–	1+ (gel)	AC negative

RBC, red blood cell; LISS, low ionic strength saline; AC, autocontrol; DAT, direct antiglobulin test; N/A, not available.

Table 2. Blood Transfusions

Patient ID	RBC units received on DARA treatment	RBC units received after DARA treatment	Hb difference per unit (g/dL)[a]
1	0	2	0.6, 0.5
2	4	0	0.8, 0.6, 0.6
3	0	1	2.2
4	1	0	1.8
5	4	0	0.3, 0.4
6	0	0	–
7	0	6	1.0, 1.3, 1.35
8	0	0	–

RBC, red blood cell; DARA, daratumumab; Hb, hemoglobin.
[a]Patients may have received multiple units per transfusion.

DISCUSSION

Role of Oncology Nurses in Mitigating DARA Interference

- DARA interference in blood typing assays intersects with many aspects of the oncology nurse's role, including patient assessment, patient and co-worker education, symptom management, and supportive care. Oncology nurses can help mitigate DARA interference by:
 - Typing and screening your patient prior to starting DARA
 - Informing the blood bank that your patient has been treated with DARA (**Figure 3**)
 - Ensuring that your patient's blood sample is identified as DARA treated
 - Double-checking transfusion standing orders to determine if your patient received DARA within the last year
 - Providing your patient's pre-DARA blood compatibility profile to the blood bank, if available
 - Asking your patient to tell his/her other health care providers that he/she has received DARA
 - Encouraging your patient to carry his/her ID card (**Figure 4**) for ≥1 year after his/her last DARA infusion
 - Reassuring your patient that transfusions can be performed safely
 - For emergency transfusions, non-crossmatched, ABO-RhD-compatible RBCs can be given

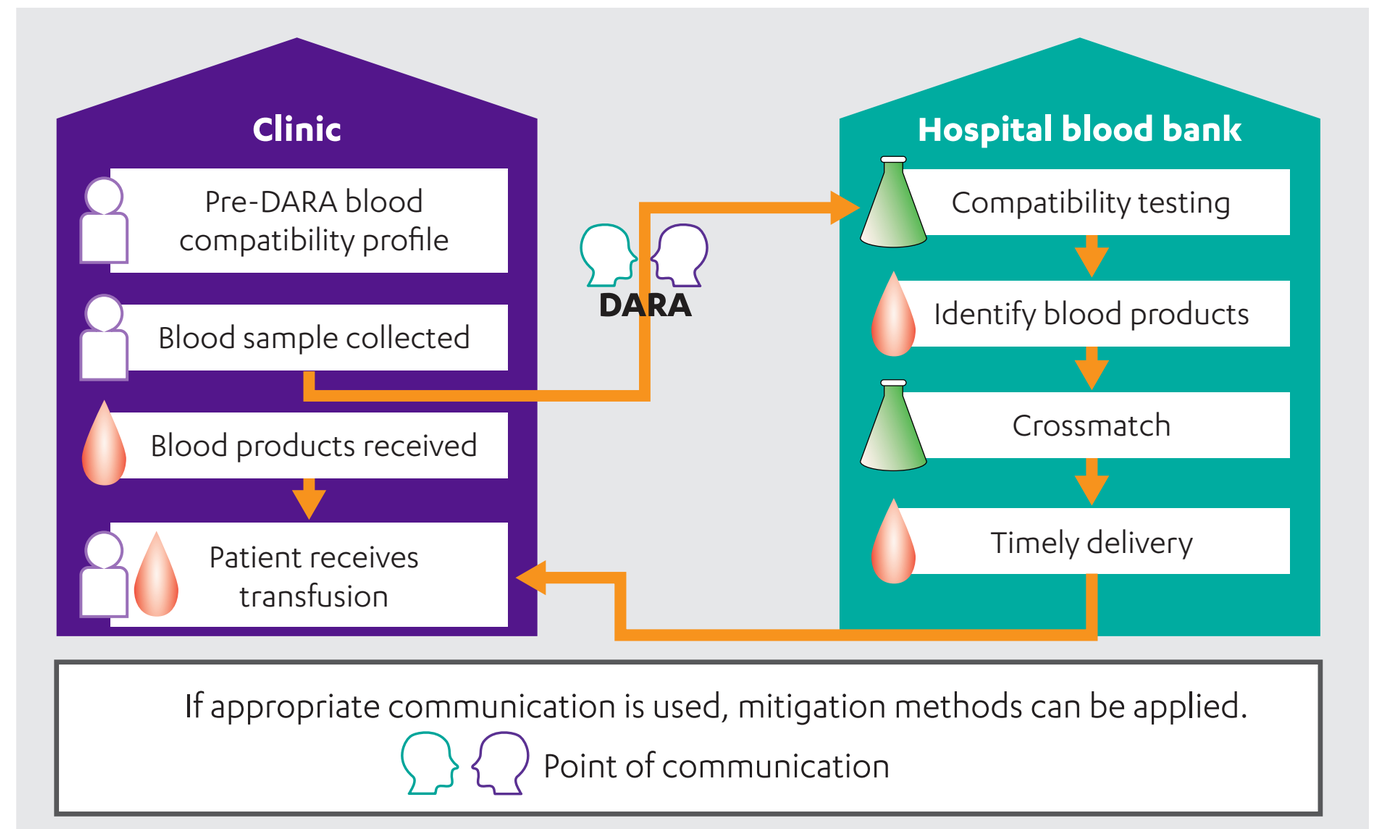
Clinic
Pre-DARA blood compatibility profile
Blood sample collected
Blood products received
Patient receives transfusion
DARA
Hospital blood bank
Compatibility testing
Identify blood products
Crossmatch
Timely delivery
If appropriate communication is used, mitigation methods can be applied.
Point of communication

Figure 3. Communication strategy for safe and timely administration of blood transfusions.

Daratumumab patients:
Provide this card to health care providers BEFORE blood transfusion

Name:______________________

I am taking the following medication:
Daratumumab, an antibody product for the treatment of multiple myeloma

Before starting daratumumab, my blood test results, collected on __ /__ /__, were:

Blood type: ☐ A ☐ B ☐ AB ☐ O ☐ Rh+ ☐ Rh–

Indirect Coombs test (antibody screen) was:

☐ Negative ☐ Positive for the following antibodies:

Figure 4. Identification card for patients receiving DARA.

CONCLUSIONS

- **Obtaining an RBC phenotype prior to initiating DARA treatment and providing phenotypically matched blood up to 1 year after the patient's last DARA infusion is one option that will prevent transfusion delays**
 - **DTT denaturation of CD38 to eliminate DARA binding to RBCs is another option**
- **At our center, blood transfusions were provided safely and without complications in concordance with observations from all 124 patients in the SIRIUS study**
- **In a patient population treated with DARA that will frequently require RBC transfusions, awareness of these findings and appropriate, customized communication between stakeholders will be important to minimize errors and transfusion delays**
- **As DARA becomes available outside of clinical trials and enters routine clinical practice, oncology nurses will play an essential and critical role in ensuring safe and timely administration of blood products for these patients by incorporating new protocols into oncology nursing practice**

REFERENCES

1. de Weers M, et al. *J Immunol*. 2011;186(3):1840-1848.
2. Lin P, et al. *Am J Clin Pathol*. 2004;121(4):482-488.
3. Santonocito AM, et al. *Leuk Res*. 2004;28(5):469-477.
4. McKeage K. *Drugs*. 2016;76(2):275-281.
5. Lokhorst HM, et al. *N Engl J Med*. 2015;373(13):1207-1219.
6. Lonial S, et al. *Lancet*. 2016. doi:10.1016/S0140-6736(15)01120-4.
7. Chapuy CI, et al. *Transfusion*. 2015;56(6 Pt 2):1545-1554.
8. Oostendorp M, et al. *Transfusion*. 2015;55(6 Pt 2):1555-1562.
9. Chari A, et al. *Blood*. 2015;126(23):3571.

ACKNOWLEDGMENTS

The authors acknowledge the patients who participated in this study and their families, as well as the study co-investigators, research nurses, and coordinators at each of the clinical sites. Editorial support was provided by Erica Chevalier-Larsen, PhD, of MedErgy, and was funded by Janssen Global Services, LLC.

An electronic version of the poster can be viewed by scanning the QR code. The QR code is intended to provide scientific information for individual reference. The PDF should not be altered or reproduced in any way.

POSTER PRESENTED AT THE ONCOLOGY NURSING SOCIETY 41ST ANNUAL CONGRESS; APRIL 28-MAY 1, 2016; SAN ANTONIO, TEXAS.